Global Influenza Hospital Surveillance Network

RESULTS FROM THE 2018/2019 INFLUENZA SEASON IN THE VALENCIA REGION OF SPAIN

Ainara Mira-Iglesias[1], Javier García-Rubio[1], F. Xavier López-Labrador[1,2], Javier Díez-Domingo[1]

[1]Fundación para el Fomento de la Investigación Sanitaria y Biomédica de la Comunitat Valenciana (FISABIO-Public Health), Valencia, Spain

[2]Consorcio de Investigación Biomédica de Epidemiología y Salud Pública (CIBERESP), Instituto de Salud Carlos III, Madrid, Spain

Site presentation

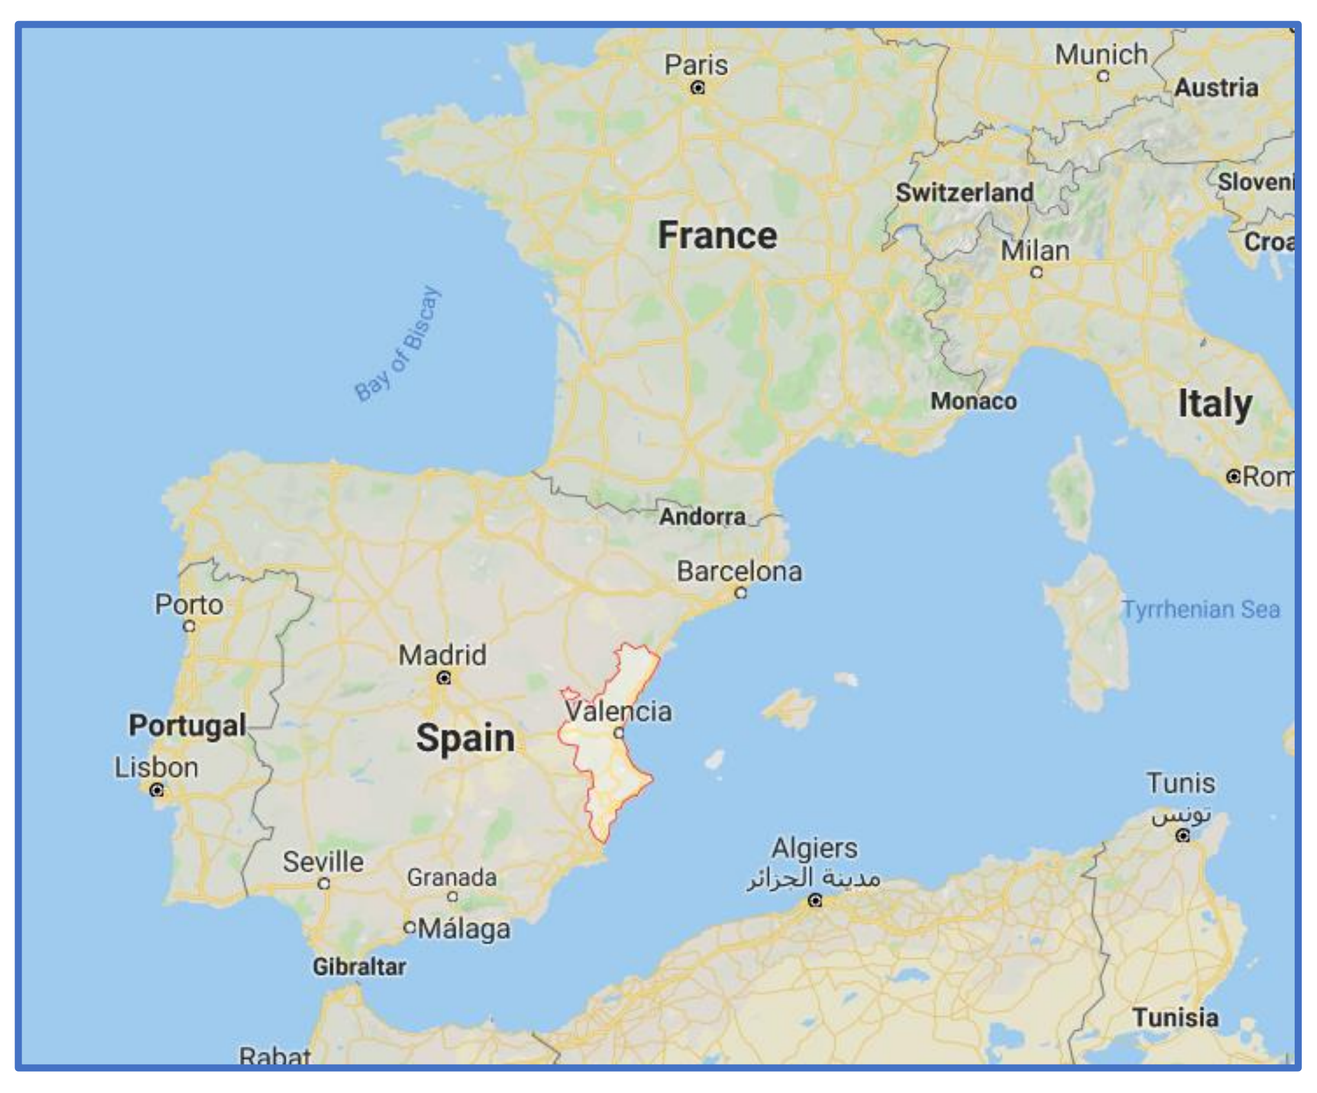

Paris
Munich
Austria
France
Switzerland
Milan
Monaco
Italy
Andorra
Barcelona
Porto
Madrid
Portugal
Spain
Valencia
Lisbon
Seville
Granada
Málaga
Gibraltar
Algiers
Tunis
Tunisia
Tyrrhenian Sea
Bay of Biscay

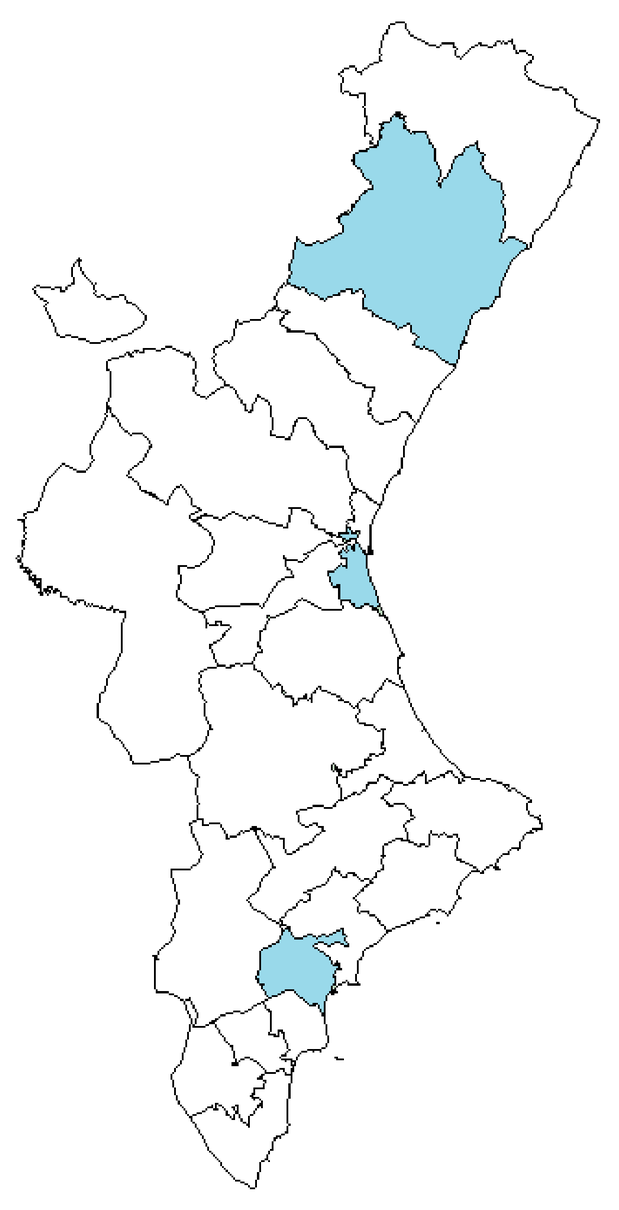

Hospital General Castellón
Population: 281,200

Hospital La Fe
Population: 285,066

Hospital Doctor Peset
Population: 278,344

Hospital General Alicante
Population: 274,122

Methods

The **Valencia Hospital Network for the Study of Influenza** (VAHNSI) conducts annually a **prospective**, **active-surveillance**, **hospital-based** study on admissions with diagnoses possibly related to influenza.

From **September to August**, all hospitalized patients were included in the study in case they:

- Gave written informed **consent**
- Were **resident** in one of the hospitals' catchment area
- Were **not institutionalized**
- Were **not** discharged from a **previous hospitalization** in the last 30 days
- Satisfied the ECDC **ILI-case definition** (required only for patients ≥5 years old)
- Were hospitalized within **7 days of the onset of symptoms**

Clinical **information** was collected through **interview** and by **medical records** review.

Information related to **vaccination** was obtained from the **Valencia Region Vaccine Information System**. Swabs were tested by real-time reverse transcription polymerase chain reaction (**RT-PCR**) in a centralized laboratory in FISABIO. Influenza Vaccine Effectiveness (IVE) was estimated by a **Bayesian logistic regression** following the **test-negative design.**

Results

Respiratory viral infections

Screened: 11,883 → Included: 4,218 → PCR-positive: 1,559

Influenza: 322; RSV: 394; HMPV: 109; PIV: 27; HRV: 609; AdV: 30; BoV: 17; CoV: 109

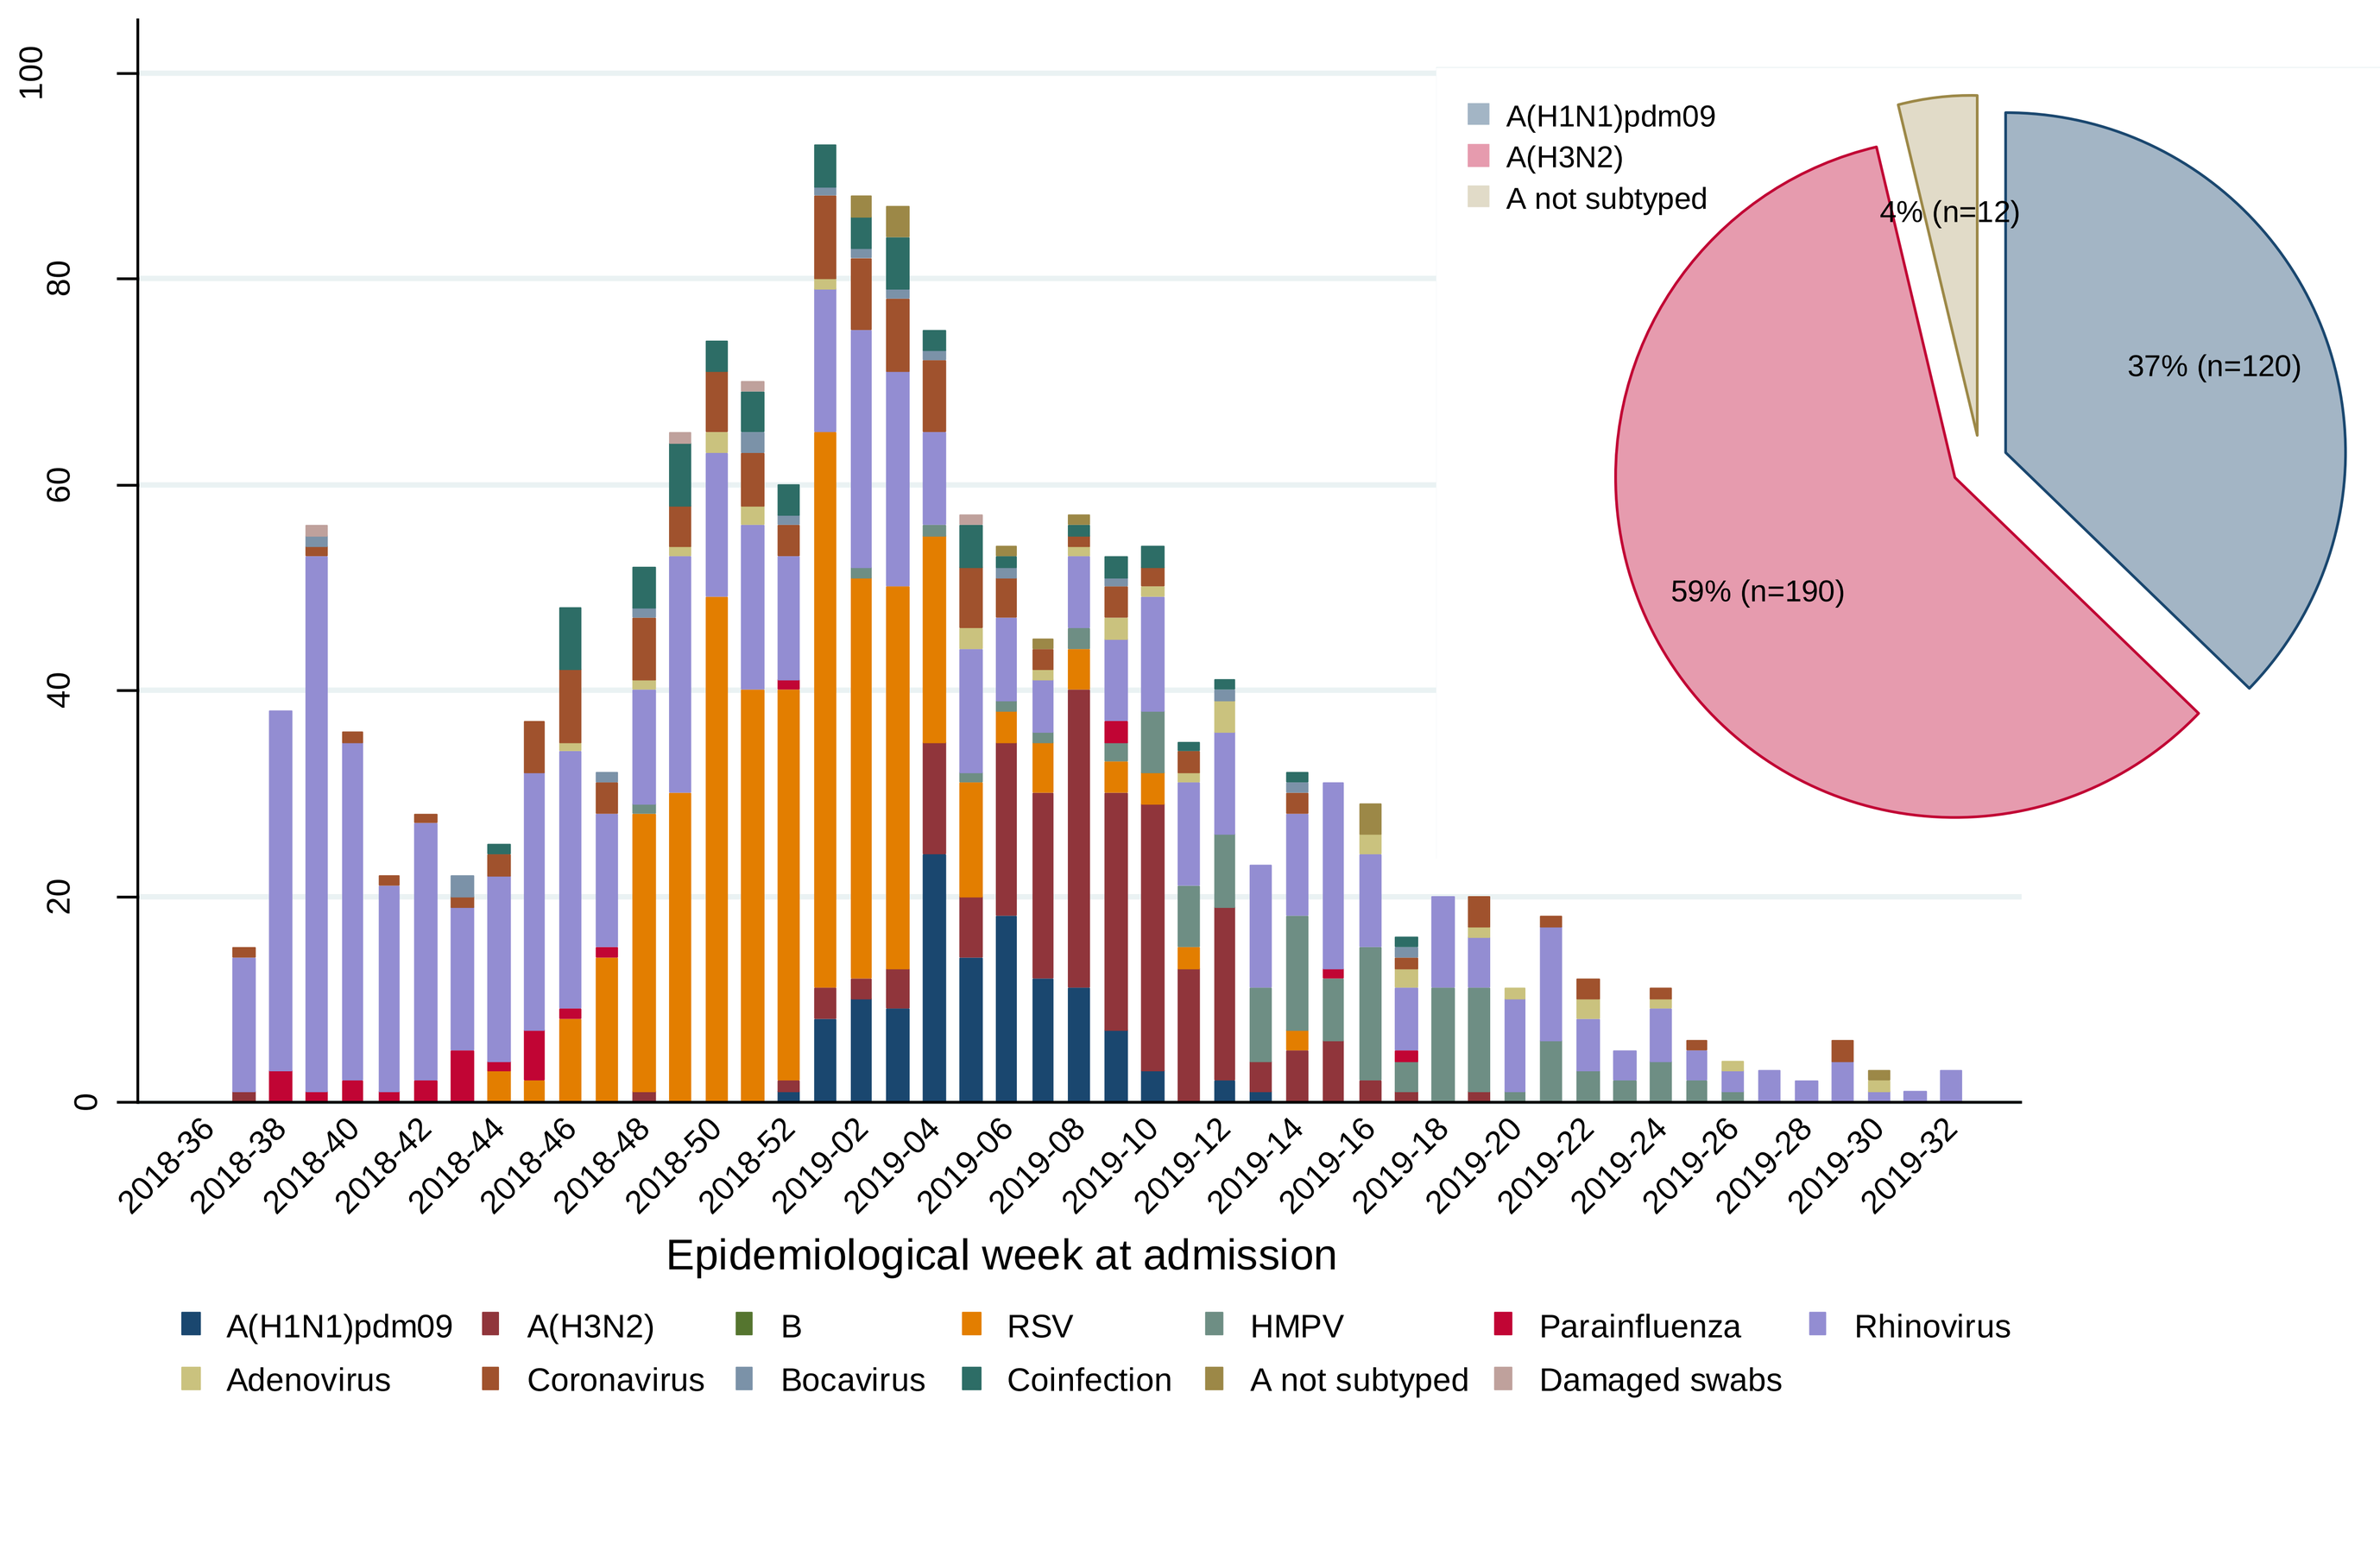

0
20
40
60
80
100
Epidemiological week at admission
A(H1N1)pdm09
A(H3N2)
B
RSV
HMPV
Parainfluenza
Rhinovirus
Adenovirus
Coronavirus
Bocavirus
Coinfection
A not subtyped
Damaged swabs
37% (n=120)
59% (n=190)
4% (n=12)

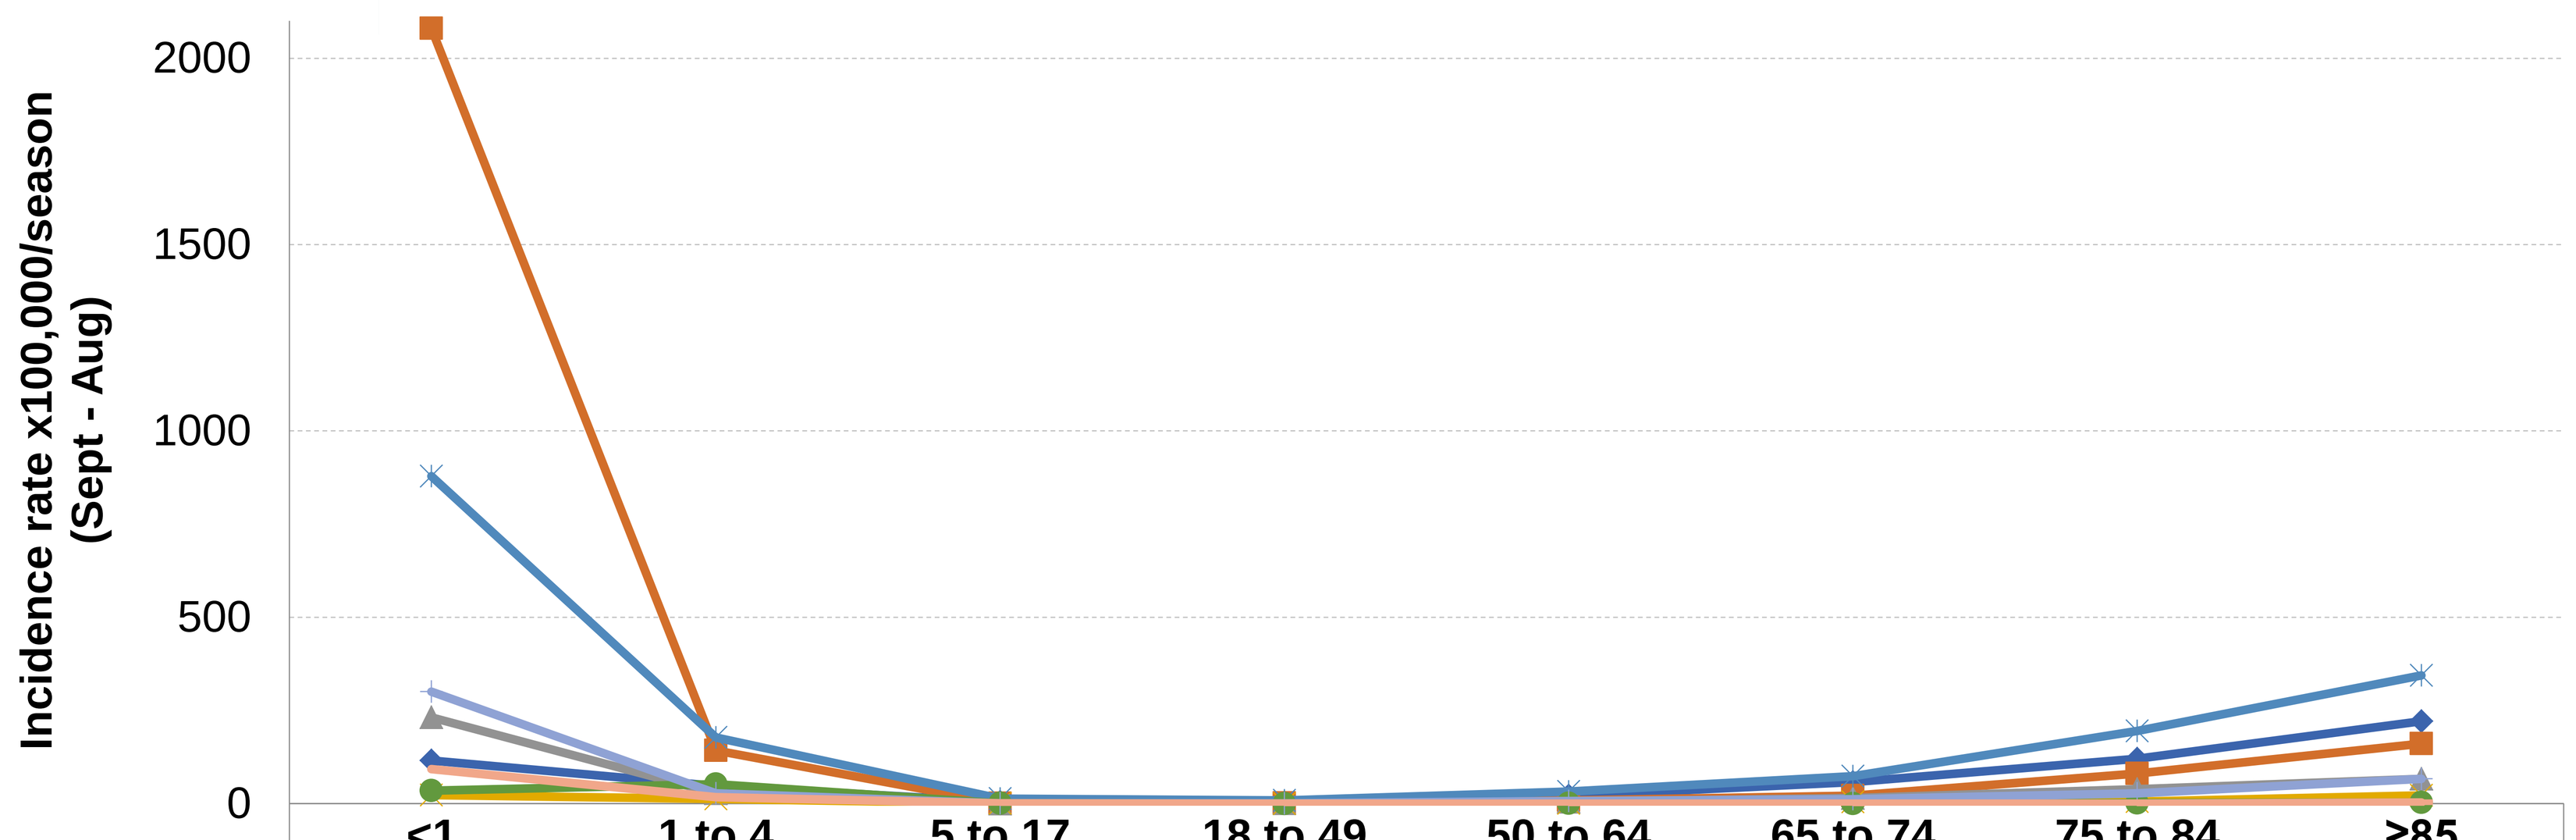

Incidence rate x100,000/season (Sept - Aug)
2000
1500
1000
500
0

	<1	1 to 4	5 to 17	18 to 49	50 to 64	65 to 74	75 to 84	≥85
Influenza	115.57	47.49	4.67	3.96	21.44	57.98	120.63	221.04
RSV	2080.20	142.46	0.67	1.25	8.75	20.84	80.42	161.05
HMPV	231.13	27.49	0.00	0.63	4.81	14.50	38.77	66.31
Parainfluenza	23.11	12.50	0.67	0.21	0.87	4.53	5.74	22.10
Rhinovirus	878.31	177.45	13.34	8.54	32.37	73.38	195.31	344.20
Adenovirus	34.67	52.49	0.67	0.21	0.87	0.00	1.44	3.16
Coronavirus	300.47	27.49	0.67	0.42	7.00	11.78	27.29	66.31
Bocavirus	92.45	17.50	0.67	0.00	0.00	0.00	0.00	3.16

Influenza vaccine effectiveness (in ≥65 years old)

IVE

- Overall 39% (95% CrI: 14, 59)
- A(H1N1)pdm09 50% (95% CrI: 10, 75)
- A(H3N2) 28% (95% CrI: -6, 55)

Genetic characterization of viruses

All the **A(H1N1)pdm09** isolated viruses corresponded to the **clade 6B.1 as the recommended A/Michigan/45/2015 vaccine virus**, but encoding several amino acid substitutions characteristic for diversification in several distinct subclades.

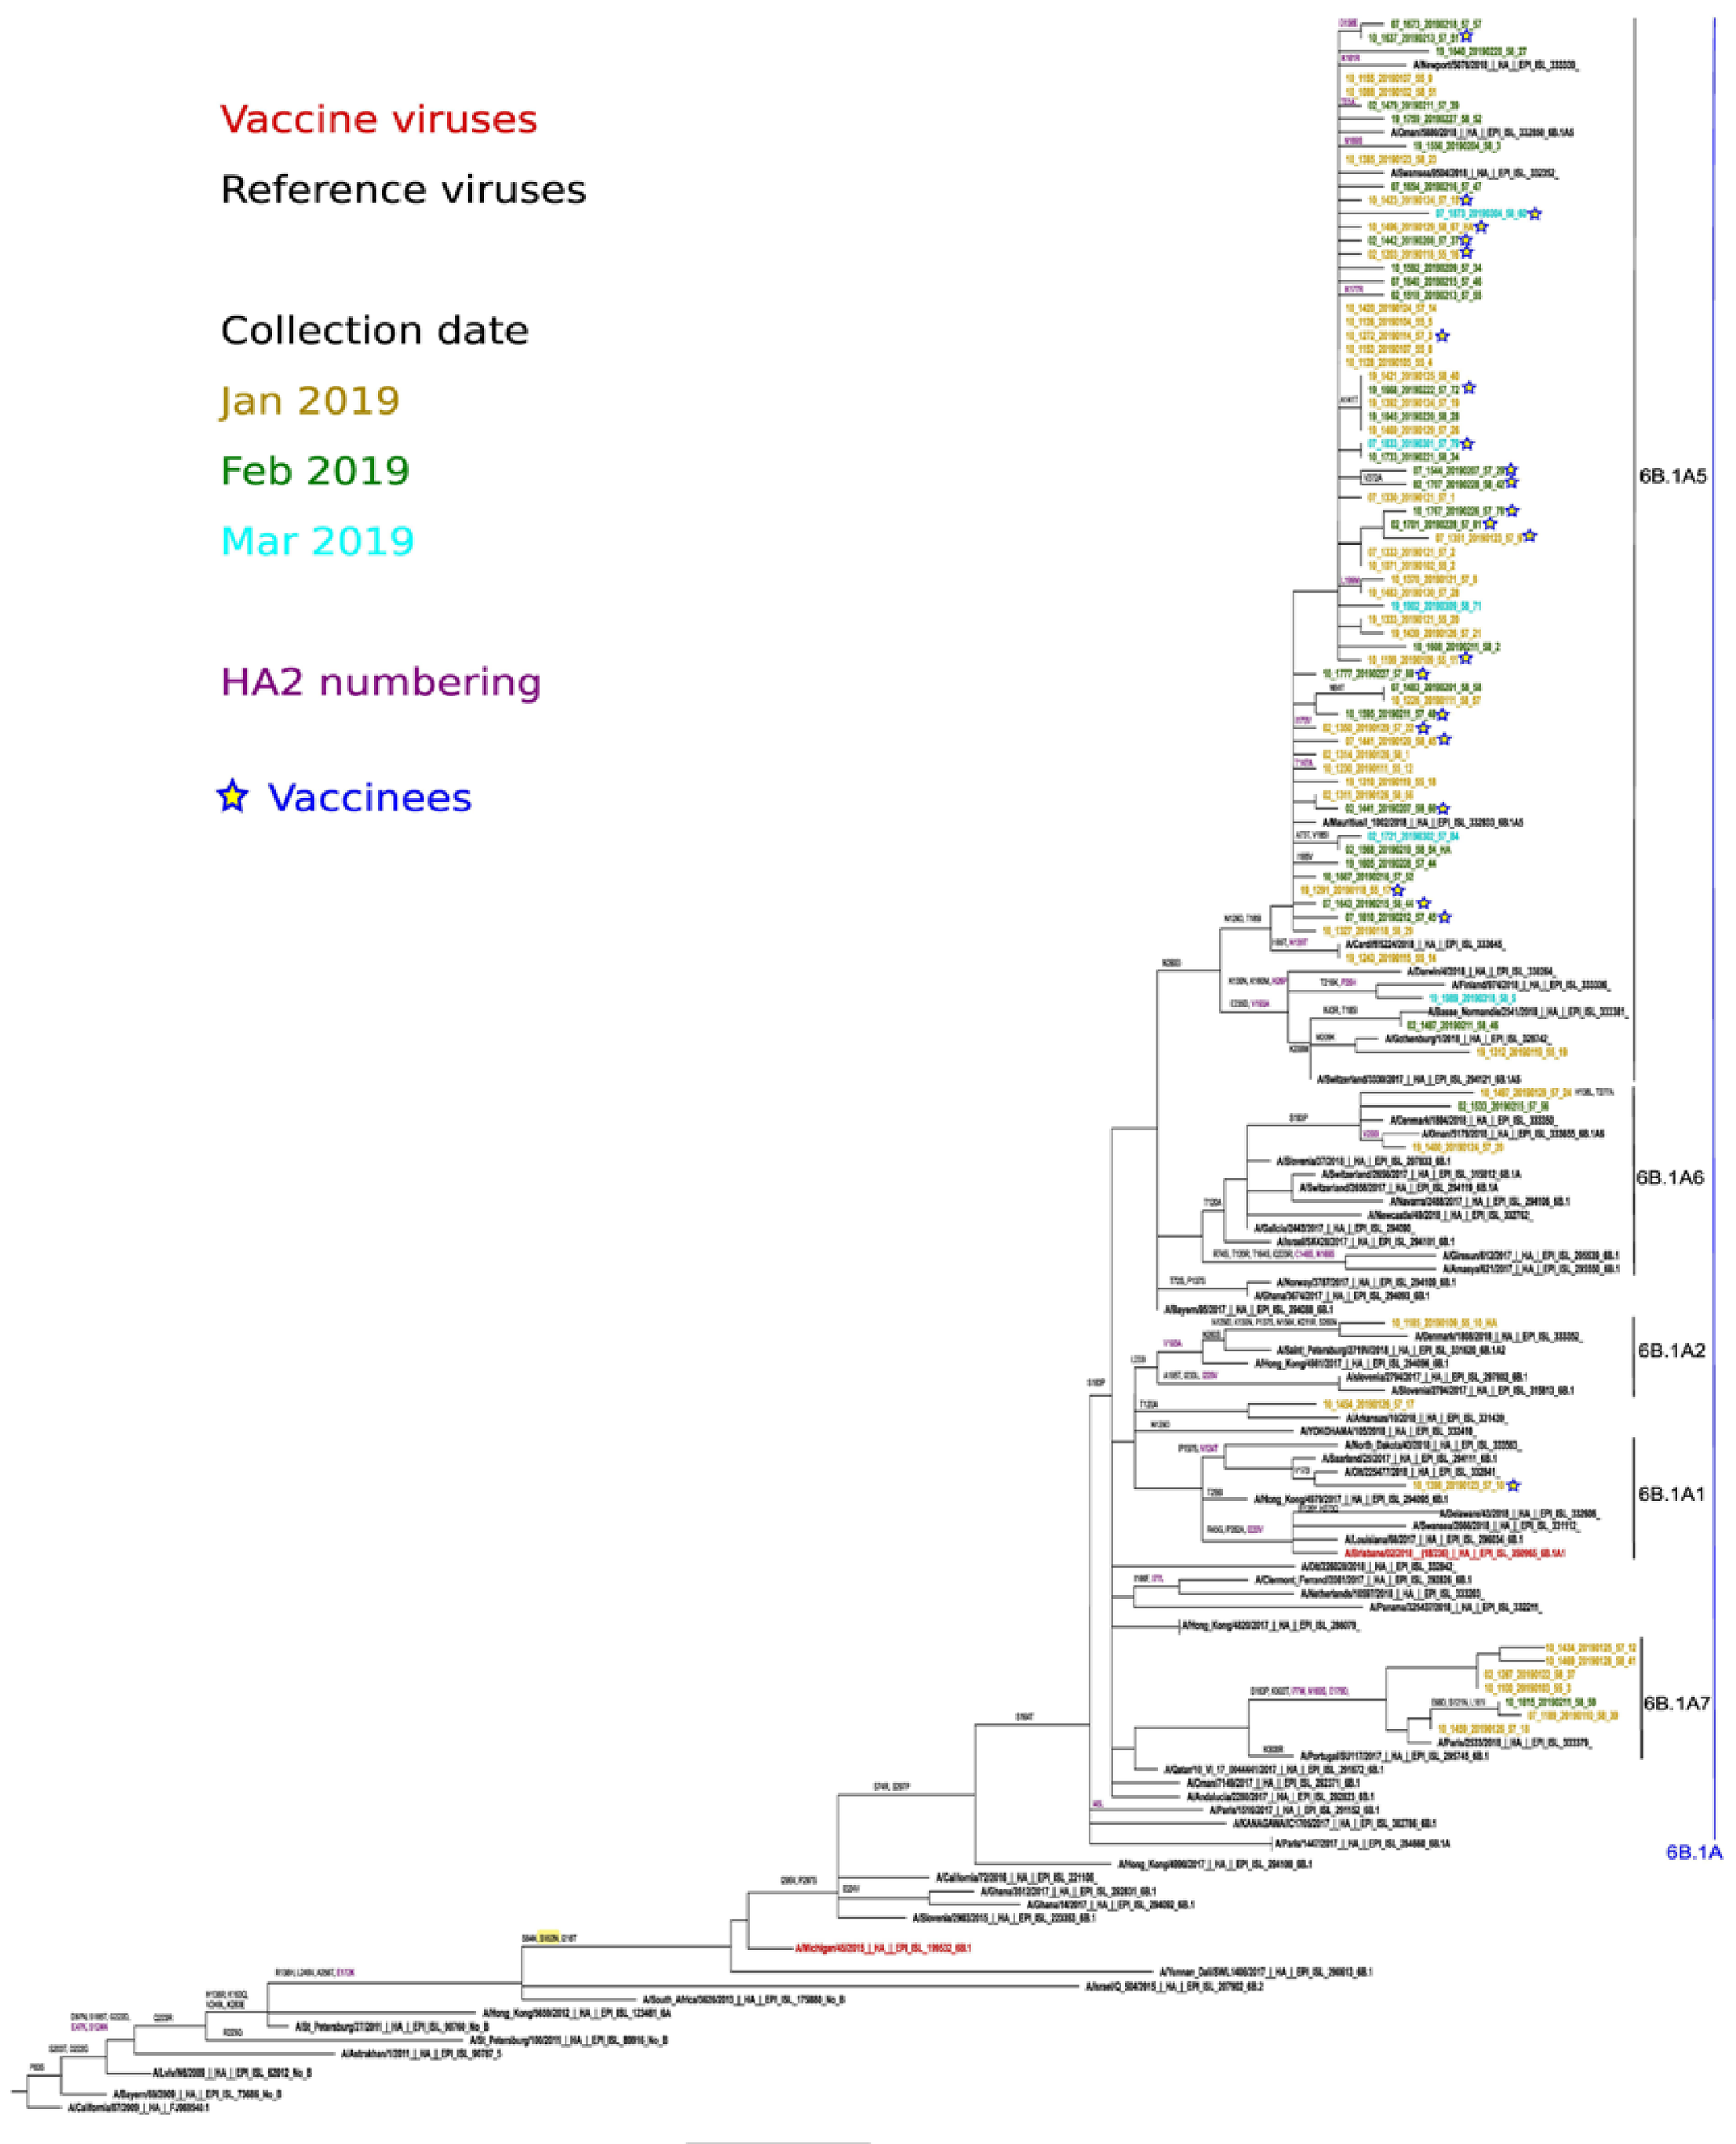

Vaccine viruses
Reference viruses
Collection date
Jan 2019
Feb 2019
Mar 2019
HA2 numbering
Vaccinees
6B.1A5
6B.1A6
6B.1A2
6B.1A1
6B.1A7
6B.1A

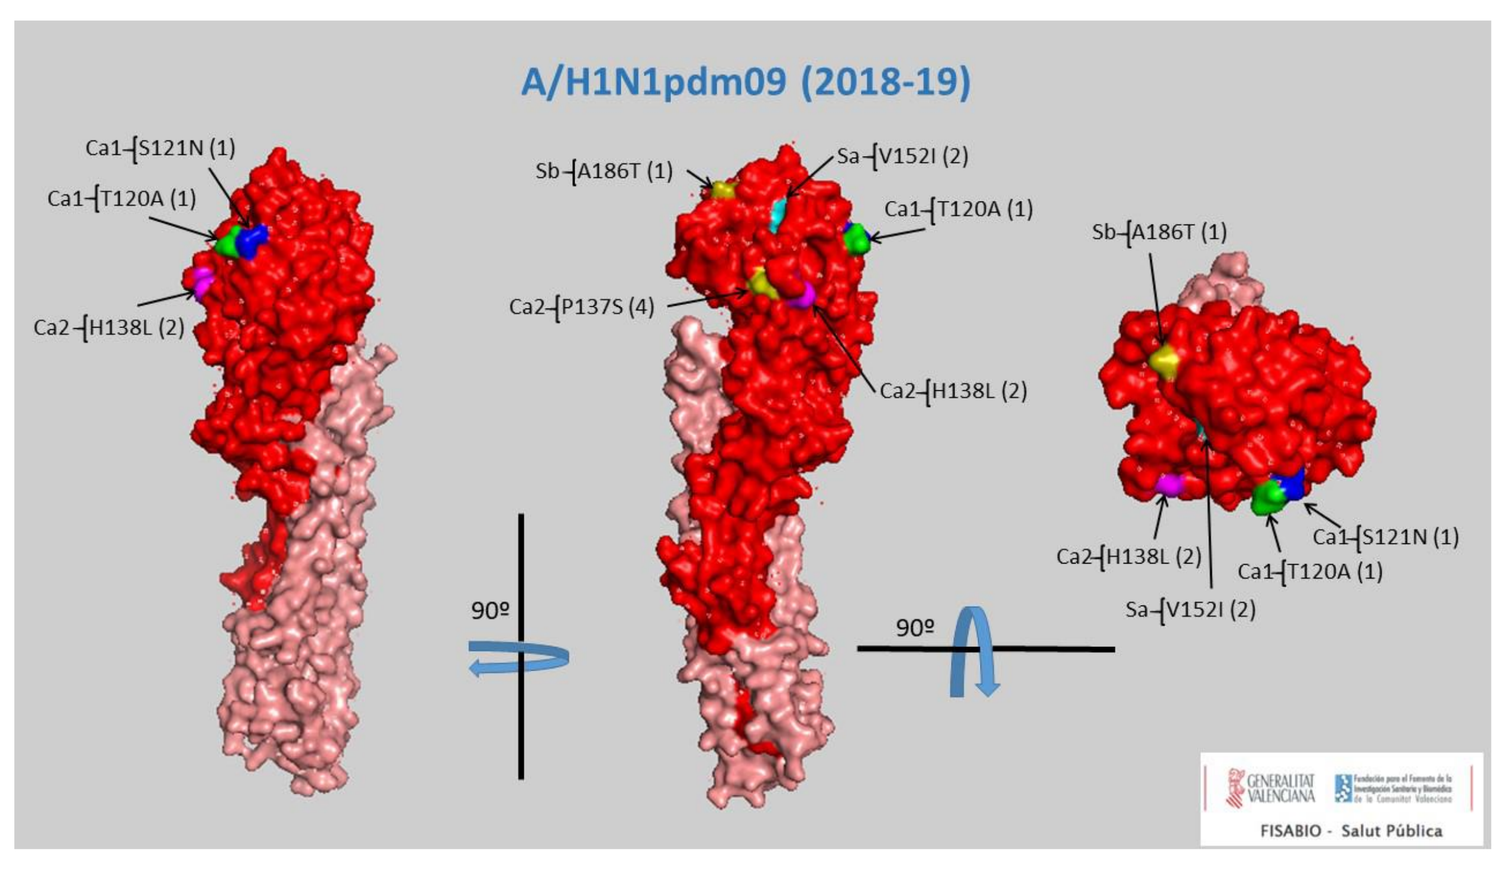

A/H1N1pdm09 (2018-19)

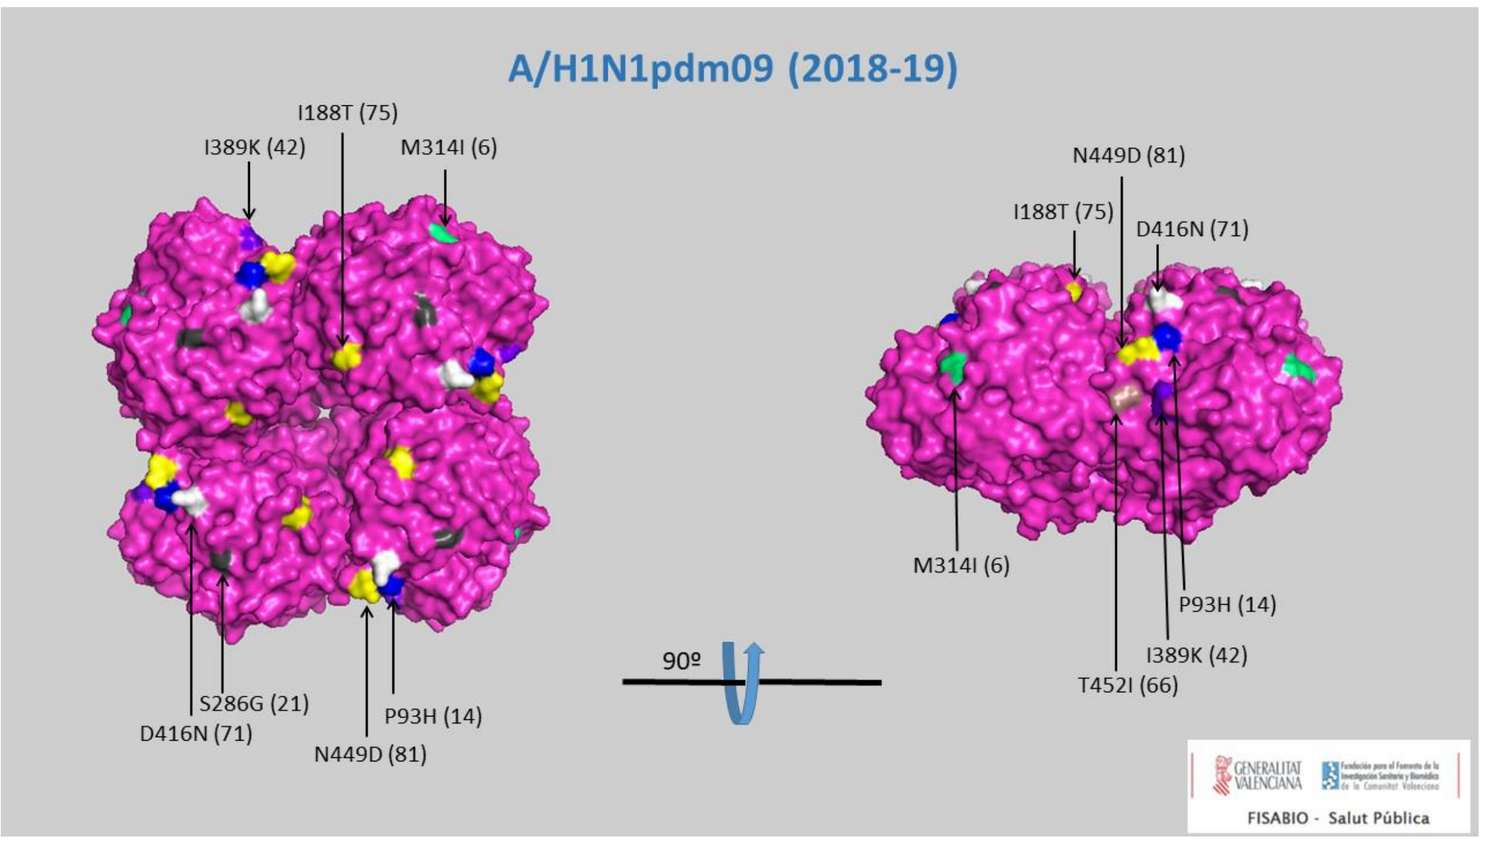

A/H1N1pdm09 (2018-19)

Phylogenetic analysis and mutational profiles for the A(H1N1)pdm09 clades and sequenced isolates.

A(H1N1)pdm09 structure showing the location and frequency of amino acid substitutions found within antigenic sites as compared with the vaccine virus.

There were similar proportions of **A(H3N2)** viruses **from subclades 3C.2a1b and 3C.3a**, which were both predominant. **None** of the sequenced isolates **corresponded to** the 2018/19 NH **clade 3C.2a1 recommended vaccine virus A/Singapore/INFIMH-16-0019/2016**.

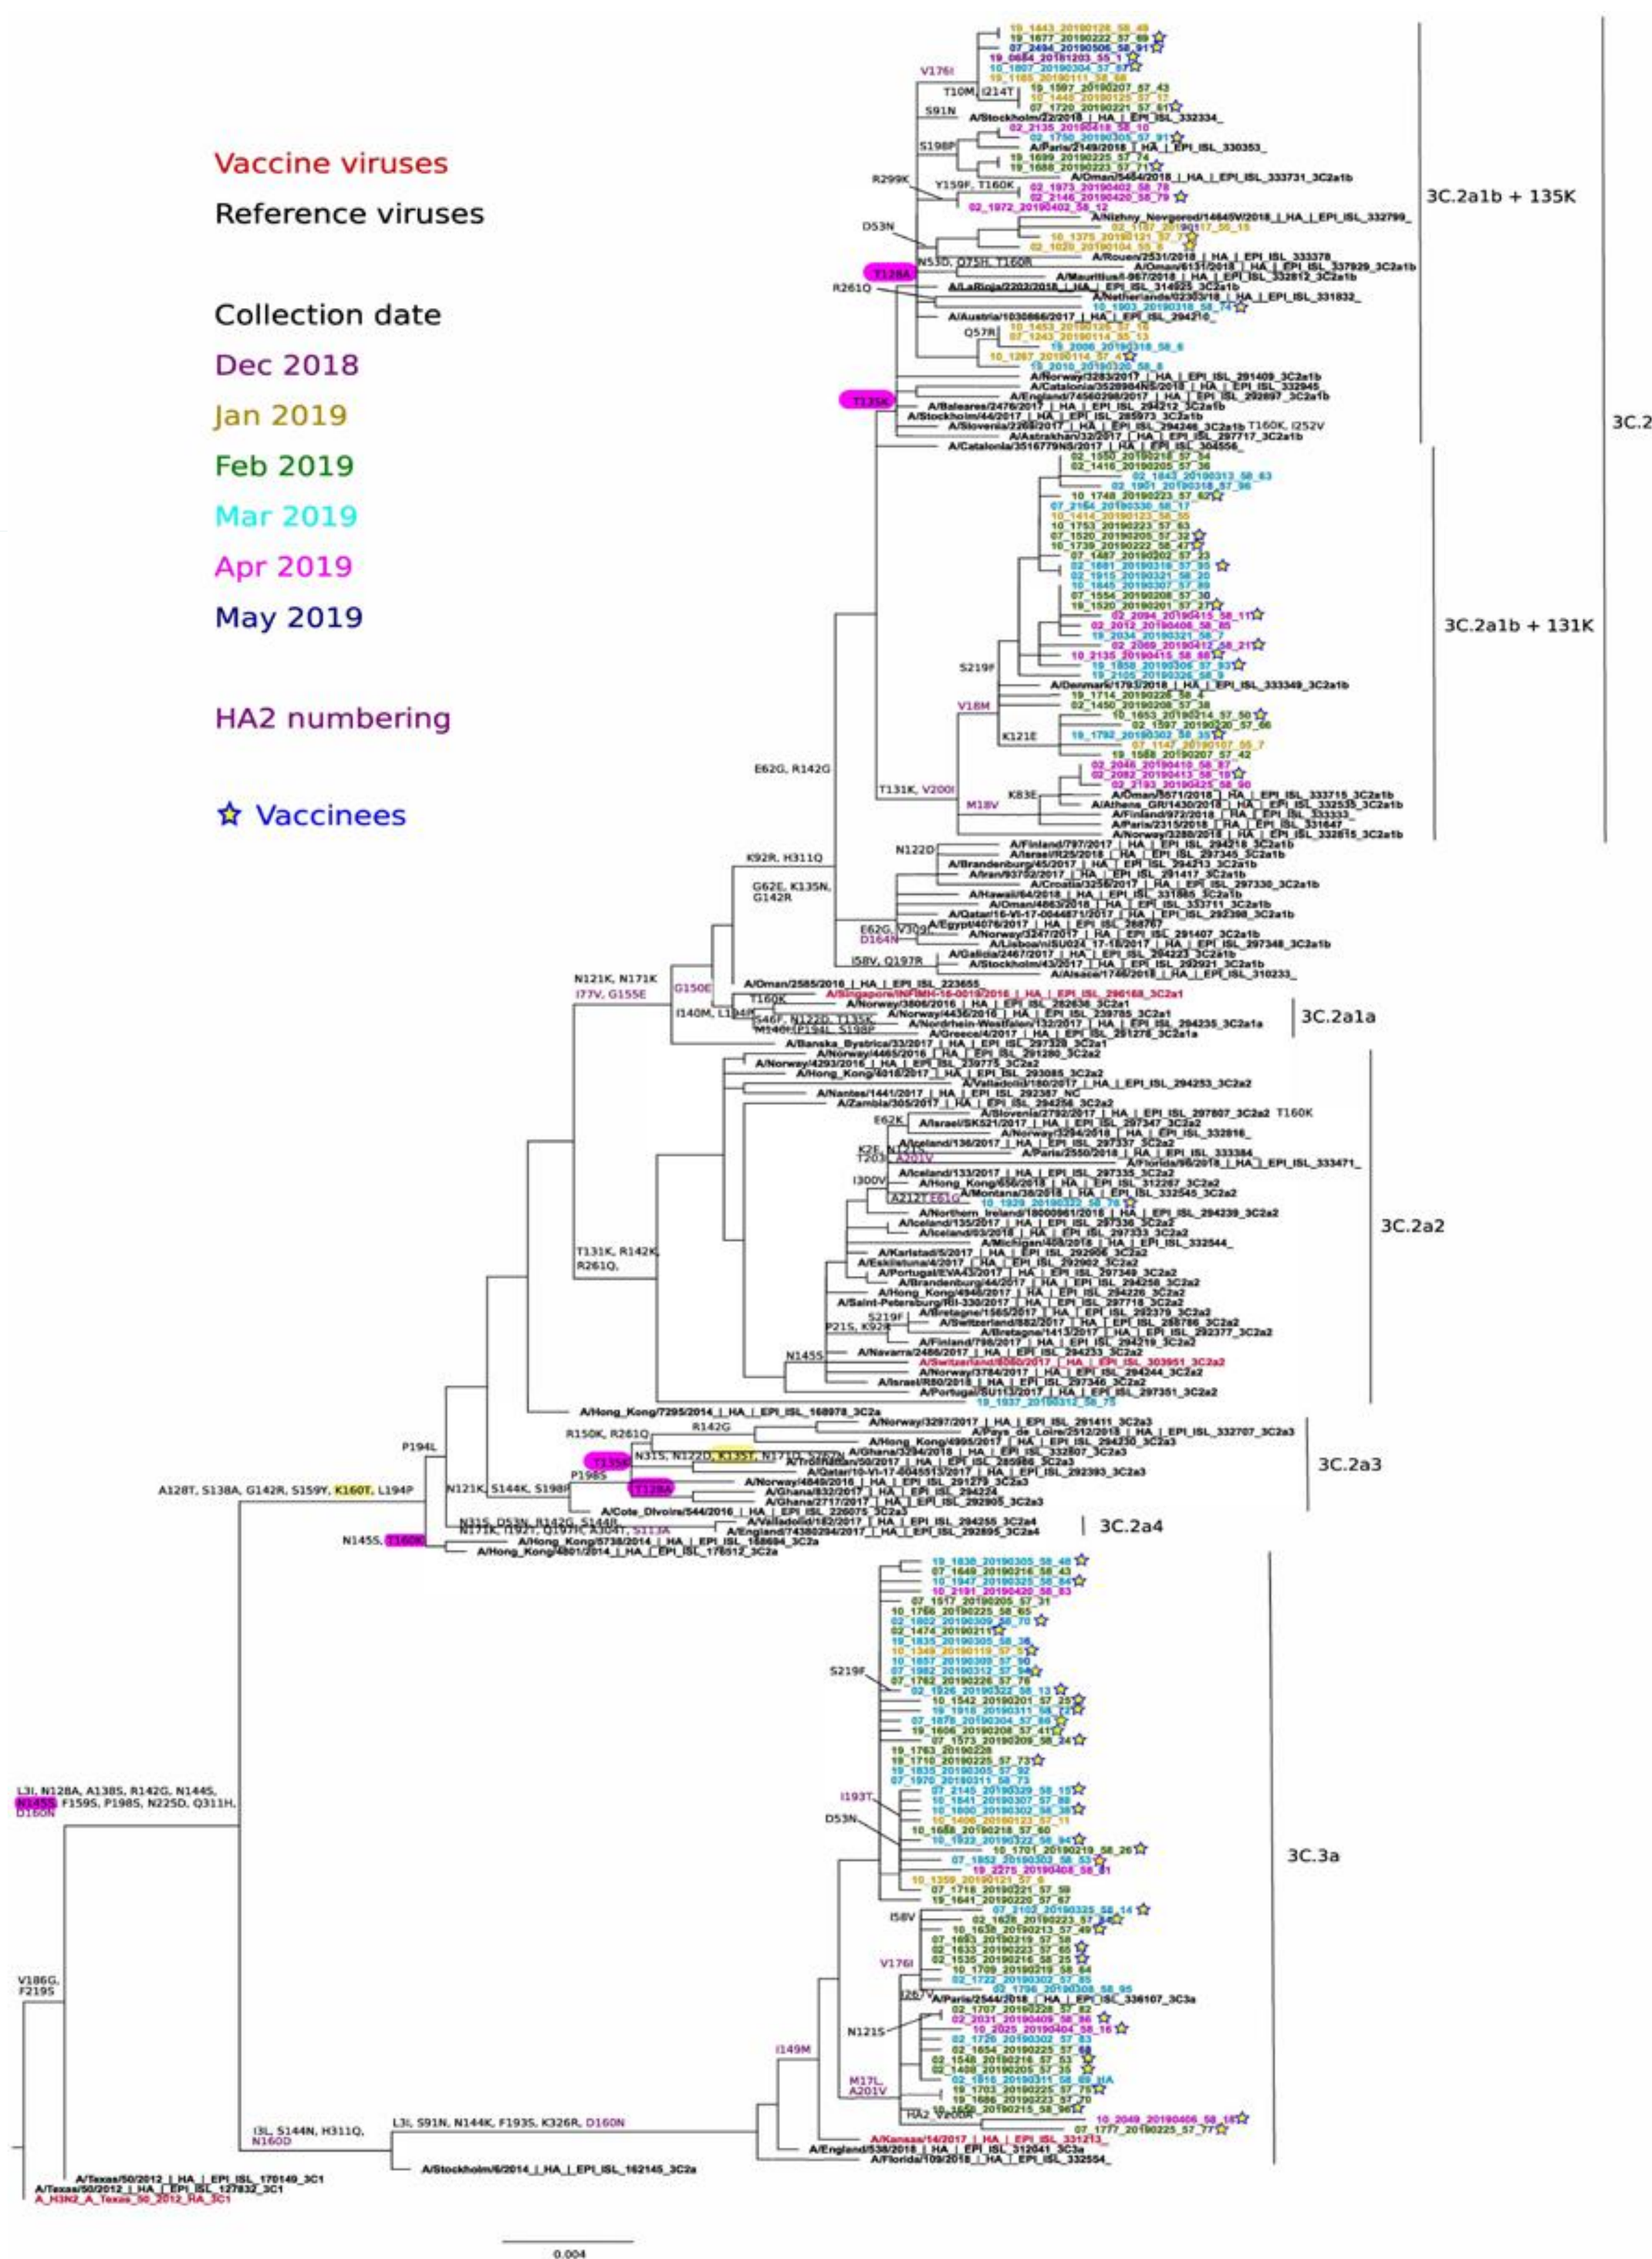

Vaccine viruses
Reference viruses
Collection date
Dec 2018
Jan 2019
Feb 2019
Mar 2019
Apr 2019
May 2019
HA2 numbering
Vaccinees
3C.2a1b + 135K
3C.2a1b
3C.2a1b + 131K
3C.2a1
3C.2a1a
3C.2a2
3C.2a3
3C.2a4
3C.3a

Phylogenetic analysis and mutational profiles for the A(H3N2) clades and sequenced isolates.

A(H3N2) structure showing the location and frequency of amino acid substitutions found within antigenic sites as compared with the vaccine virus.

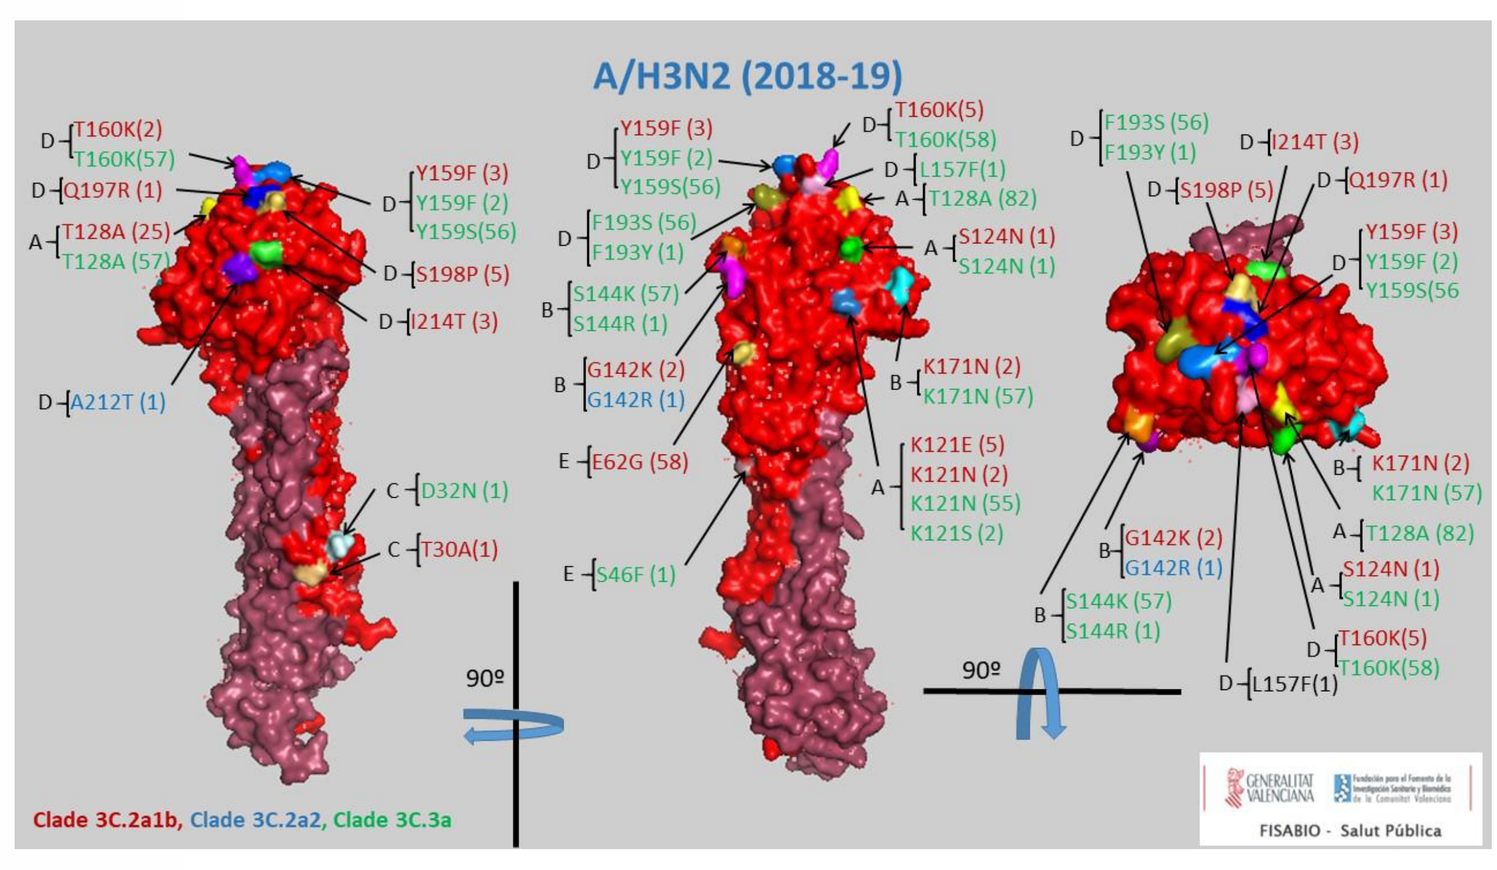

A/H3N2 (2018-19)
Clade 3C.2a1b, Clade 3C.2a2, Clade 3C.3a

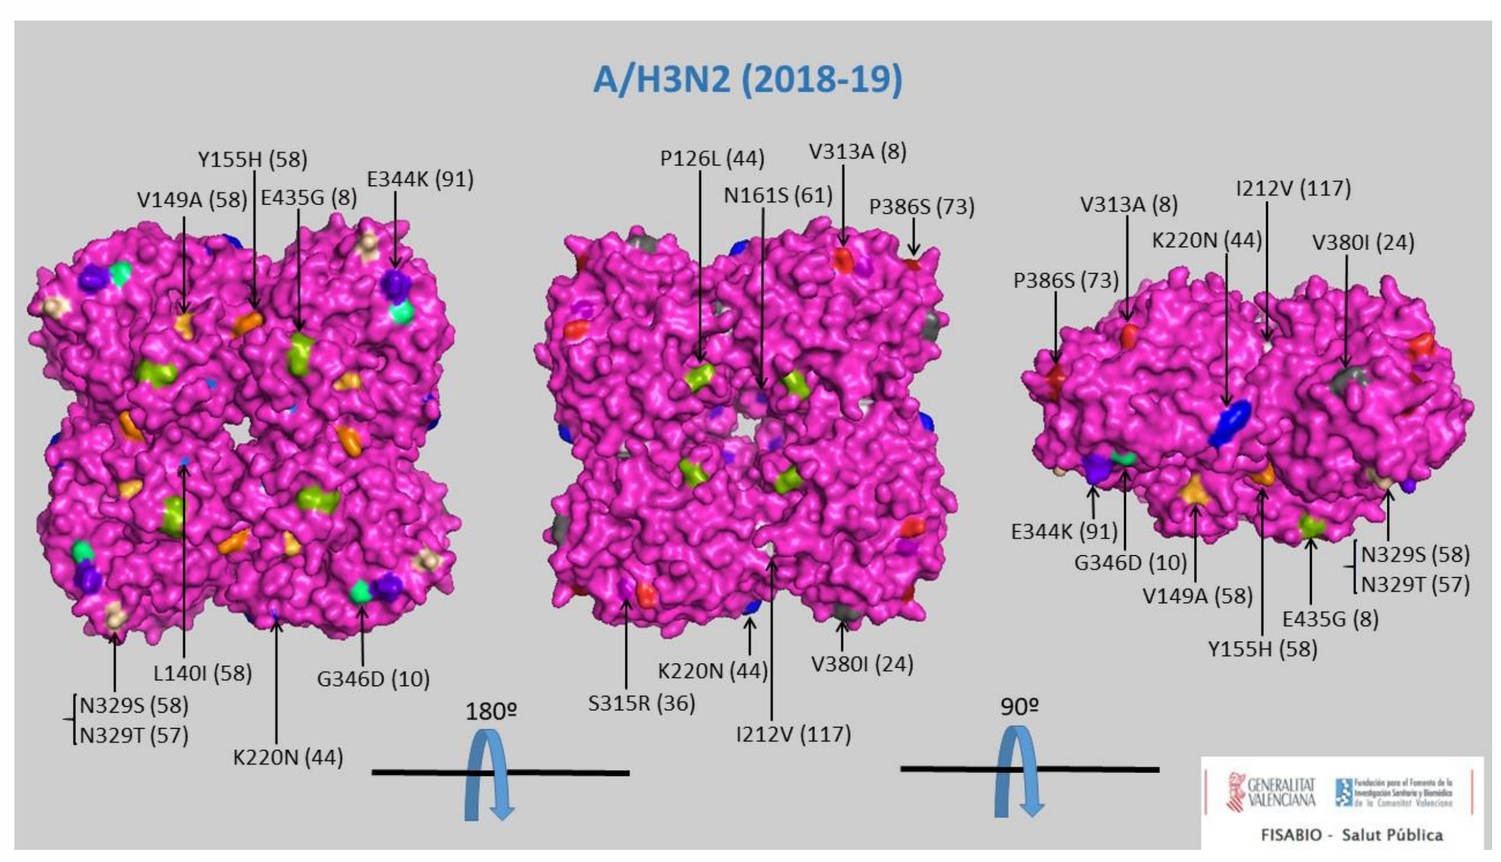

A/H3N2 (2018-19)

Key aspects & challenges

Circulation of influenza A(H1N1)pdm09 and A(H3N2) with no B cases.

The influenza peak was reached at week 2019-08.

Highest influenza incidence rate was detected in 85+.

Highest RSV and RhV incidence rates were detected in <1.

IVE was moderate against overall influenza and A(H1N1)pdm09 with no significant protection against A(H3N2).

All the A(H1N1)pdm09 isolated viruses belonged to the vaccine virus clade. None of the A(H3N2) isolated viruses corresponded to the vaccine virus recommendation.

Contact: Javier Díez-Domingo (diez_jav@gva.es). Head of the Vaccine Research Department, FISABIO-Public Health, Valencia, Spain.

Funding: This study was funded by FISABIO-Public Health, CIBER-ESP (ISCIII) and Sanofi Pasteur.